Phase I Study of a Liposomal Carrier (CPX-351) Containing a Synergistic, Fixed Molar Ratio of Cytarabine (Ara-C) and Daunorubicin (DNR) in Advanced Leukemias

E. Feldman, J. Lancet, J.E. Kolitz, E. Asatiani, L. Mayer, A. Louie

Weill Medical College of Cornell University and New York Presbyterian Hospital, New York, NY; H. Lee Moffitt Cancer Center and Research Institute, Tampa, FL; North Shore University Hospital, Manhasset, NY; Lombardi Cancer Center, Georgetown University Medical Center, Washington DC; Celator Pharmaceuticals, Inc., Princeton, NJ

Introduction

In vitro the efficacy of some chemotherapy doublets has been shown to be sensitive to the molar ratio of the combination. In vivo maintenance of a particular molar ratio that maximizes synergy and avoids antagonism is not possible with current formulations due to differences of drug distribution and elimination for each agent. This obstacle to enhancing antitumor efficacy has been overcome by encapsulating chemotherapies within drug carriers (liposomes or nanoparticles) which control drug elimination, enabling delivery of synergistic drug ratios to tumor cells (CombiPlex® technology). Applying this "ratiometric" approach has enabled translation of observed in vitro synergy to the in vivo setting resulting in fixed molar ratio formulations with potent therapeutic activity. CPX-351 is a liposomal formulation of Cytarabine and Daunorubicin encapsulated at the 5:1 molar ratio, a ratio that is synergistic and avoids antagonism across multiple leukemic and solid tumor cell lines. A phase I dose escalating study was conducted with CPX-351 in patients with relapsed or refractory acute leukemia.

Methods

Patients with advanced AML, ALL and high risk MDS were eligible. CPX-351 was given by 90 minute infusion on Days 1, 3 and 5 of each induction course. Second inductions were permitted with evidence of antileukemic effect and persistence of leukemia in a Day 14 bone marrow. The starting dose was 3 u/m^2 (1 u = 1 mg cytarabine and 0.44 mg daunorubicin) and doses were doubled with each single patient cohort until evidence of drug effect was observed. Thereafter, 3-patient cohorts and 33% dose increments were continued until limiting toxicities signaled the end of dose escalation. Pharmacokinetic samples were collected on Days 1, 3 and 5 of the first induction course and analyzed for cytarabine, daunorubicin, Ara-U and daunorubicinol by LC-MS/MS. The cut-off date for data collection was September 30, 2008.

Demographics and Disposition

Thirty-seven patients were recruited in 10 cohorts during the study's dose escalation phase. Maximum Tolerated Dose (MTD) was declared at the cohort 9 dose level (101u/m^2). To confirm tolerability and collect preliminary information on efficacy, the protocol was amended to include an additional 10 patients at the MTD bringing the total number of patients to 47.

Treatment outcome: 55% had persistent leukemia, 23% achieved CR, 17% were discontinued following adverse events, 4.3% received stem cell transplant (one after CR and one after PR) and 2% were lost to follow up.

Patient Demographics and Disposition:

		Cohort 1-3	Cohort 4	Cohort 5	Cohort 6	Cohort 7	Cohort 8	Cohort 9	Cohort 10	TOTAL	
	CPX-351 dose(u/m^2)	3,6,12	24	32	43	57	76	101(MTD)	134		
		n=4	n=4	n=4	n=4	n=3	n=3	n=19	n=6	n=47	%
Gender	Male	3	3	4	0	2	3	12	4	31	66
	Female	1	1	0	4	1	0	7	2	16	34
Age (yr)	Median	44.3	63.5	64.5	74	61	60	57	64.5	62	
	≥60yrs	2	2	3	3	2	2	8	4	26	55
	<60yrs	2	2	1	1	1	1	11	2	21	45
Race	White	3	4	3	4	1	2	17	6	40	85
	Black/African American	1	0	0	0	0	1	1	0	3	6
	Asian	0	0	1	0	2	0	1	0	4	9
ECOG	0	2	0	1	3	1	1	10	4	22	47
	1	1	4	3	0	2	2	8	2	22	47
	2	1	0	0	1	0	0	1	0	3	6
Diagnosis	AML	1	4	4	2	3	3	18	6	41	87
	Secondary AML	1	0	0	1	0	0	1	0	3	6
	ALL	2	0	0	1	0	0	0	0	3	6
Response to Last Line of Therapy											
	None	4	2	3	2	3	2	7	4	27	57
	CR	0	2	1	2	0	1	12	2	20	43
Reason Off Study											
	(Serious) Adverse Event	0	1	1	1	0	0	4	1	8	17
	Persistent Leukemia	4	3	2	1	3	3	7	3	26	55
	Bone Marrow Transplant	0	0	0	0	0	0	2*	0	2*	4
	Discretion of Investigator or Sponsor	0	0	0	0	0	0	0	0	0	0
	Lost to Follow-up	0	0	0	0	0	0	1	0	1	2
	Complete Remission	0	0	1(CRp)	2	0	0	6**	2	11**	23

* - One of the BMT patients achieved a PR
** - One of the CR patients transferred for BMT and is counted twice.

Conflict of Interest Disclosure

L. Mayer and A. Louie are employees of Celator Pharmaceuticals and own stock or stock options in the company. There are no other relevant conflicts of interest to disclose.

Safety Profile of CPX-351

Table 2:
Non-hematological High Grade Adverse Events

	Cohorts 5-8		Cohort 9		Cohort 10		TOTAL	
CPX-351 dose(u/m^2)	32-76		101(MTD)		134			
	n=11		n=19		n=6		n=36	%
Adverse Event	Gr. 3	Gr. 4	Gr. 3	Gr. 4	Gr. 3	Gr. 4		
Infections								
Febrile Neutropenia	3	0	8	2	4	0	17	47
Bacteremia	1	0	7	0	1	0	9	25
Pneumonia	1	0	7	0	0	0	8	22
Sepsis/Septic Shock	0	0	4	0	0	1	5	14
UTI	0	0	0	0	1	0	1	3
Cellulitis	0	0	1	0	0	0	1	3
Clostridial Infection	0	0	1	0	1	0	2	6
Bleeding Events								
Intracerebral	1	0	0	0	0	0	1	3
GI bleeding	1	0	0	0	0	0	1	3
Mucosal/Skin	1	0	2	1	0	0	4	11
CNS								
Aphasia	0	0	0	0	0	1	1	3
Convulsion	0	0	0	0	1	0	1	3
Depressed Level of Consciousness	0	0	0	0	1	0	1	3
Metabolic								
Hypokalemia	0	0	2	1	1	1	5	14
Metabolic Acidosis	0	0	1	0	0	0	1	3
Hypophosphatemia	1	0	0	0	0	0	1	3
Skin								
Rash	1	0	1	0	1	0	3	8
Cardiovascular								
LV Dysfunction/Orthopnea	0	0	1	0	1	0	2	6
Pericardial Effusion	0	0	1	0	0	0	1	3
Atrial Fibrillation	0	0	0	0	1	0	1	3
Hypertensive Crisis/Hypertension	0	0	0	0	1	1	2	6
Respiratory								
Dyspnea	1	0	2	0	0	0	3	8
ARDS	0	0	1	0	0	0	1	3
Hypoxia	1	0	0	0	1	0	2	6
Aspiration	0	0	0	0	1	0	1	3
Cough	1	0	0	0	0	0	1	3
Gastrointestinal								
Nausea/vomiting	0	0	0	0	1	0	1	3
Diarrhea	0	0	1	0	0	0	1	3
Mucositis	0	0	0	1	1	0	2	6
Hepatic								
Hyperbilirubinemia	0	0	1	0	0	0	1	3
↑AST/↑ALT	0	0	1	0	0	0	1	3
Renal								
Acute Renal Failure	1	0	1	0	0	0	2	6
General								
Fatigue	0	0	1	0	0	0	1	3
Lithium toxicity	0	0	1	0	0	0	1	3
Pyrexia	1	0	0	0	2	0	3	8
Asthenia/Weakness	0	0	0	0	1	0	1	3

- 29 deaths occurred during the study with 19 (66%) due to persistent leukemia and 4 (14%) due to infections, 2 (7%)-respiratory failure, 1 (3%)-cerebral hemorrhage, 1 (3%) status epilepticus, 1 (3%)-diarrhea, 1 (3%)-unknown;
- At 101 u/m^2 3 deaths (16%) occurred by day 30, one death (5%) by day 60 and 3 deaths (16%) after day 60;
- Days 31-60 mortality was 15% (7 patients), days 61-90 was 11% (5 patients) and long term mortality beyond day 90 stood at 23% (11 patients).
- 34 SAEs occurred in 30 patients with nearly half caused by potential or documented infections including febrile neutropenia (10 events), fever (5), pneumonia (5), bacteremia (2), sepsis (1), cellulitis (1) and death secondary to infection (1) or to bleeding episodes including mucosal bleeding (2), epistaxis (1), GI bleeding (1) and hemorrhagic stroke (1);
- Grade 3-4 non-hematological AEs are shown for the dose levels 5-10 in Table 2, Grade 3 and 4 AEs, especially infections, are dose dependent;
- Dose limiting toxicities occurred in cohort 10 (134 u/m^2) and included single patients with hypertensive crisis (Grade 4), LV dysfunction (Grade 3) and persistent thrombocytopenia requiring platelet transfusion support beyond Day 56;
- Major GI toxicity was uncommon with 1 patient having Grade 3 N+V (134 u/m^2), 1 patient with Grade 5 diarrhea (101 u/m^2) and single patients having Grade 3 (134 u/m^2) and Grade 4 (101 u/m^2) mucositis, no other major GI toxicities were observed;
- At 134u/m^2 1 patient had shortness of breath (LVEF52%→29%) in the setting of enterococcal bacteremia after 546mg/m^2 of lifetime cumulative DNR exposure, a second patient who received 101u/m^2 x 2 inductions (lifetime cumulative DNR=966mg/m^2) had orthopnea, both patients recovered. Among the 17 patients documented to have received at least 400mg/m^2 of anthracycline only 2, described above, developed clinically apparent cardiac dysfunction.

Table 3:
Mortality by period

	Cohort 1-3	Cohort 4	Cohort 5	Cohort 6	Cohort 7	Cohort 8	Cohort 9	Cohort 10	TOTAL	
CPX-351 dose(u/m^2)	3,6,12	24	32	43	57	76	101(MTD)	134		
	n=4	n=4	n=4	n=4	n=3	n=3	n=19	n=6	n=47	%
Deaths									6	
≤7 days	0	0	0	0	0	0	0	0	0	0
8-30 days	2	0	0	1	0	0	3	0	6	13

Efficacy of CPX-351

Eleven CR/CRp were observed during the study:
- 1 CR among 3 ALL patients
- 10 CRs (9 CR + 1 CRp) among 44 AML patients:
 - **2nd – 7th salvage: 3 CR/22(14%)**
 - 2nd salvage: 1 CR/12(8%)
 - 3rd salvage: 1 CR + 1 CRp/7(29%)
 - 4th – 7th salvage: 0 CR/3pts
 - **1st salvage: 7CR/22(32%)**
 - primary refractory: 1 CR/5 (20%)
 - 1st relapse ≤60 yrs: 4 CR/7 (57%)
 - 1st relapse >60 yrs: 2 CR/10 (20%)

Cytogenetics for AML patients who achieved CR
- Normal (4 pts), <3 abnormalities (4 pts), >3 abnormalities (2 pt)
- All first salvage CRs had received Cytarabine + Anthracycline for initial induction treatment
- CRs were achieved with doses as low as 32 u/m^2 after 2 inductions and as low as 43 u/m^2 after single induction

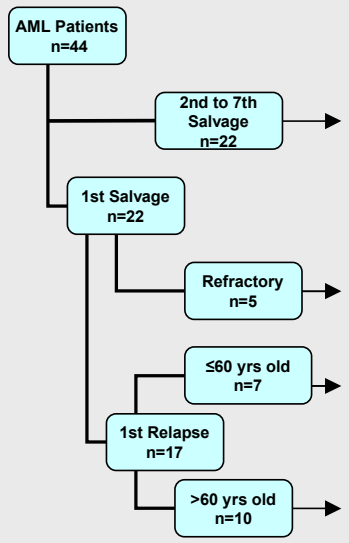

AML Patients n=44
2nd to 7th Salvage n=22
1st Salvage n=22
Refractory n=5
≤60 yrs old n=7
1st Relapse n=17
>60 yrs old n=10

Table 4: Complete Remissions Summary in AML patients, (ALL patients CR – not shown here)

Patient	Age	Cytogenetic abnormalities	Prior Treatment/CR duration (months)	CPX-351 as Salvage #	Dose (u/m^2)	No. Consolidations	Aplasia duration (days)
02-012	37	3 abnormalities t(X;2)(q26;q34), inv(7)(p13q22), t(11;15)(p13;q24)	HSCT (23mos) HSCT (11mos)	2nd	101 x 1	0	47
03-006	55	1 abnormality +8	7+3 (2mos) 5+2 (9mos) HiDAC (10mos)	3rd	32 x 2	0	94
03-008	74	3 abnormalities t(X;17)(q?22;p13), -9, del(20)(q11.2q13.3)	Zarnestra/CTEP (no response) Arsenic/AraC (7mos) Lintuzumab (no response)	3rd	43 x 1	1	35
02-011	62	1 abnormality del(5)(q13;q33)	Remlivid/7+3 (no response)	1st	134 x 1	0	131
03-017	36	normal	7+3/5+2 (9mos)	1st	101 x 1	1	57
03-019	56	normal	7+3/HiDAC (20mos)	1st	101 x 1	1	47
03-011	60	1 abnormality t(11;19)(q23;p13.1)	7+3 (3mos)	1st	76 x 1	1	35
03-018	59	normal	7+3/HiDAC (12mos)	1st	101 x 1	1	42
03-015	67	1 abnormality del(5)(q?13;q?22)	Idarubicin/Cytarabine (56mos)	1st	134 x 1	1	42
03-022	72	normal	7+3 (8mos)	1st	101 x 1	1	41

Pharmacokinetics of CPX-351

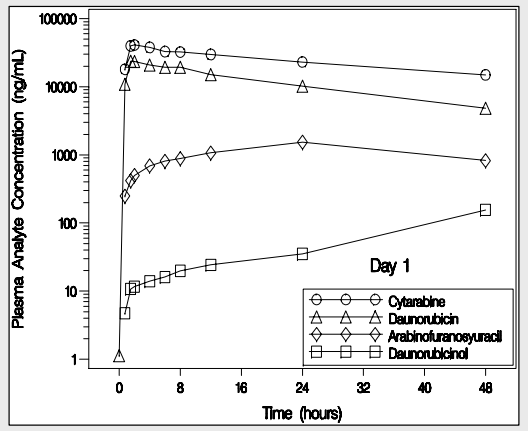

Plasma Analyte Concentration (ng/mL)
Time (hours)
Day 1
Cytarabine
Daunorubicin
Arabinofuranosyuracil
Daunorubicinol

Figure 1. Day 1 Mean Plasma Cytarabine, Daunorubicin, Ara-U and Daunorubicinol concentrations, Time 0-48hr (101u/m^2, 90 min infusion)

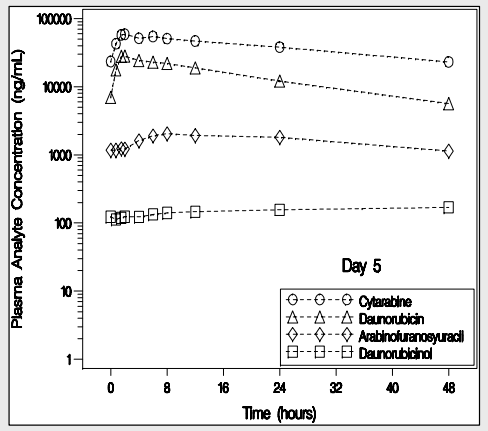

Plasma Analyte Concentration (ng/mL)
Time (hours)
Day 5
Cytarabine
Daunorubicin
Arabinofuranosyuracil
Daunorubicinol

Figure 2. Day 5 Mean Plasma Cytarabine, Daunorubicin, Ara-U and Daunorubicinol concentrations, Time 0-48hr (101u/m^2, 90 min infusion)

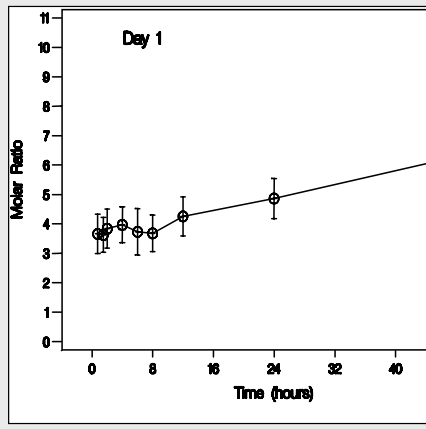

Day 1
Molar Ratio
Time (hours)

Figure 3. Mean (SD) Plasma Cytarabine:Daunorubicin, Molar Ratios Following Day 1 Administration, Time 0-48 hr (101u/m^2, 90 min infusion)

Table 5: Individual and Summary of Cytarabine and Daunorubicin

Pharmacokinetic Parameters on Day 5 Following Multiple-Dose Administration of 101units/m^2 CPX-351

	Cmax (ng/mL)	Tmax (hr)	AUC(0-tau) (ng*hr/mL)	$t_{½}$ (hr)
Cytarabine				
n	13	13	13	13
Median	55,800	2	1,487,638	31.1
Mean	64,608	3.02	1,851,089	36.9
SD	23,230	2.25	934,523	24.5
Daunorubicin				
n	13	13	13	13
Median	29,200	2	633,579	21.9
Mean	30,185	1.87	666,640	25.2
SD	6,198	0.74	209,198	11.6
SD	103	15.6	4,030	23.3

- Cytarabine and daunorubicin are bioavailable.
- The molar ratio of cytarabine:daunorubicin is maintained near a 5:1 range for >24 hours.
- CPX-351 exhibits first order elimination kinetics and minimal early-phase distribution.
- Compared to Day 1, the Day 5 plasma elimination kinetics of both drugs and their metabolites are comparable but absolute concentrations are elevated and drug levels remain detectable for 7 days after the Day 5 dose.
- Median $t_{½}$ for cytarabine was 31.1 hours and 21.9 hours for daunorubicin, suggesting that cytotoxic drug levels are likely to persist into the second week of induction (Days 8-14).
- PK data establishes the bioavailability of cytarabine and daunorubicin, the markedly prolonged half life for both drugs and their metabolites and the maintenance of the intended molar ratio for periods greater than 24 hours.

Discussion

CPX-351 is a liposomal fixed ratio formulation of Cytarabine and Daunorubicin that seeks to enhance efficacy by exposing tumor cells to synergistic molar ratios ("ratiometric" dosing). The drugs are encapsulated at a 5:1 molar ratio and animal model studies revealed potent therapeutic activity with acceptable toxicity. Similarly, this phase I study demonstrated acceptable toxicity up to 101u/m^2. The most frequent severe adverse events were those related to febrile neutropenia and documented infections. Adverse events were consistent with those expected from conventional cytarabine and daunorubicin treatment. Encouraging evidence of anti-leukemic activity was observed in ALL (1CR/3) and AML (9CR+1CRp/44). Although 7 of the 10 AML CRs occured at the two highest dose levels, complete remissions also occurred at 32u/m^2 (CRp) and at 43 and 76u/m^2. CRs occurred in patients after multiple relapses, in patients >70yrs, in patients with 3 cytogenetic abnormalities and in patients relapsing after prior cytarabine + anthracycline inductions. The phase I data suggests that CPX-351 may be useful in the relapsed setting and may be competitive with conventional 7+3 treatment in the first line setting.

CONCLUSIONS

- ✓ Dose limiting toxicities were observed at 134u/m^2.
- ✓ The recommended dose for phase II study (MTD) is 101u/m^2 by 90 minute infusion given on Days 1, 3 and 5.
- ✓ The most frequent and severe adverse events were those related to potential or documented infections including fever, febrile neutropenia, bacteremia, pneumonia and sepsis.
- ✓ PK data established cytarabine and daunorubicin bioavailability and confirmed that liposomal encapsulation results in prolonged half life for both drugs and their metabolites.
- ✓ Complete remissions were observed in 10 (9CR + 1 CRp) subjects with AML and 1 with ALL.
- ✓ Phase II studies are being initiated in newly diagnosed AML patients aged 60-75 and in 1st relapse AML patients aged 18-60.

Presented at the 50th ASH Annual Meeting & Exposition, 6-9 December 2008, San Francisco, CA